Development of Intranasal Carbetocin (LV-101) for the Treatment of Hyperphagia and Behavioral Symptoms in Prader-Willi Syndrome

Elisabeth M. Dykens,[1] Jennifer Miller,[2] Moris Angulo,[3] Elizabeth Roof,[1] Kathryn King,[4] and Sara Cotter[4]

[1] Kennedy Center for Research on Human Development, Vanderbilt University, Nashville, TN; [2] Division of Pediatric Endocrinology, University of Florida, College of Medicine, Gainesville, FL; [3] Department of Pediatrics, Winthrop University Hospital, Mineola, NY; [4] Levo Therapeutics, Inc., Skokie, IL

#839

Background

PWS

Prader-Willi syndrome (PWS) is a rare, life-threatening neurodevelopmental genetic disorder. There is evidence of a dysfunctional oxytocin system in PWS mediating hyperphagia and possibly other behavioral symptoms such as obsessions, compulsions, and anxiety. There is no currently approved treatment for hyperphagia or behavioral symptoms in PWS.

Carbetocin

This Phase 2 study evaluated the safety and efficacy of intranasal carbetocin in subjects with PWS.

Carbetocin is a synthetic analogue of oxytocin that was specifically designed to increase selectivity for the oxytocin receptor versus vasopressin receptors, providing a wider therapeutic window than oxytocin. Intravenously administered carbetocin is approved in over 70 countries for uterine atony and excessive bleeding during Caesarean section. More than 10 million patients have been exposed to intravenous carbetocin. In 2017, Ferring Pharmaceuticals licensed intranasal carbetocin to Levo Therapeutics.

Objective

To assess the effect of intranasal carbetocin on hyperphagia and other behavioral symptoms in subjects with PWS.

Design/Methods

Study design

Prospective, double-blinded, placebo-controlled, Phase 2 study

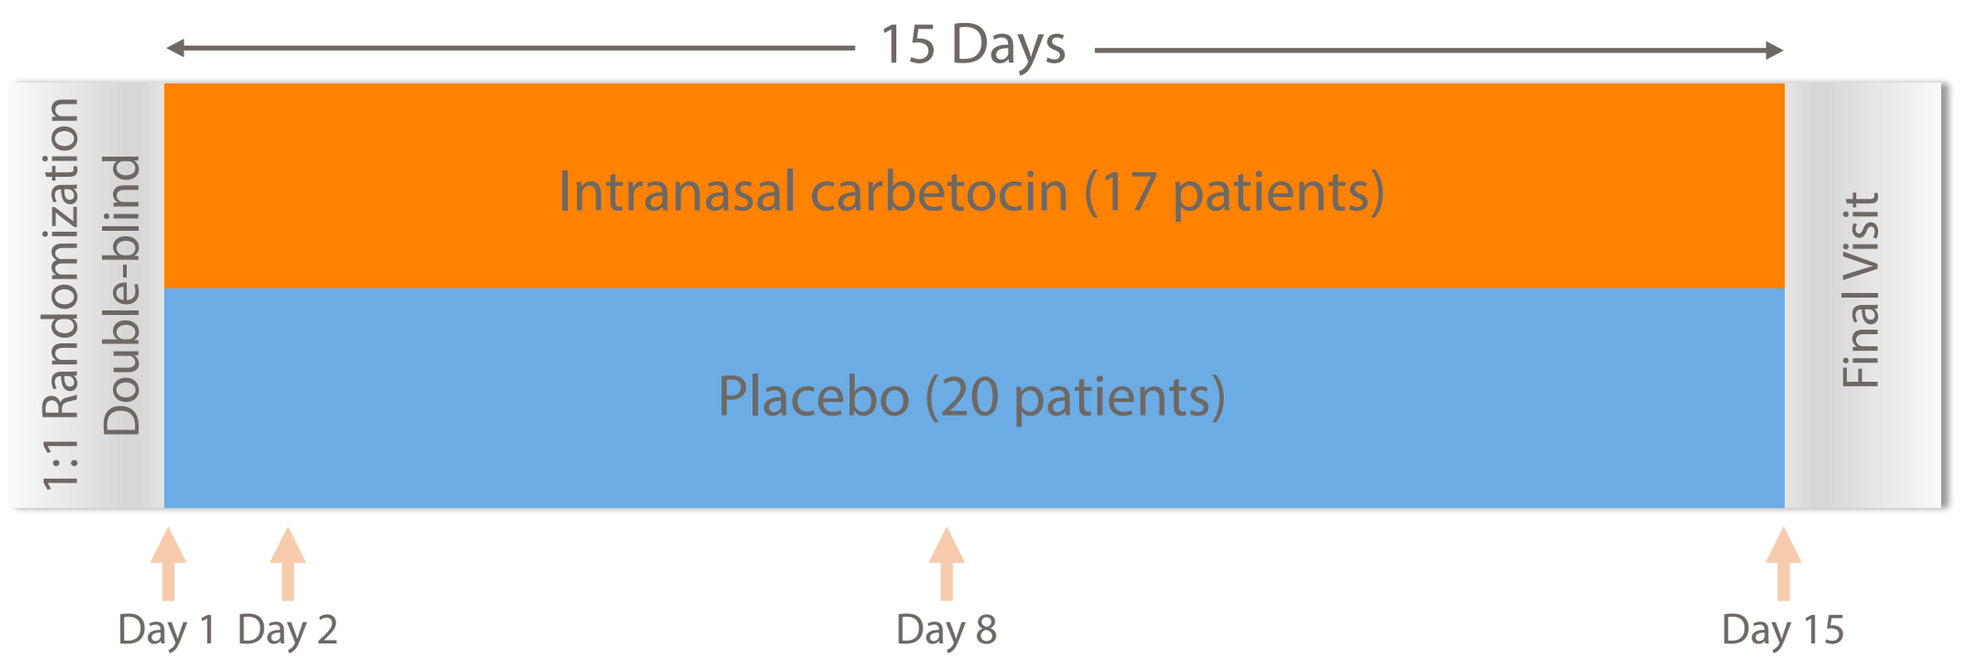
15 Days
1:1 Randomization
Double-blind
Intranasal carbetocin (17 patients)
Placebo (20 patients)
Final Visit
Day 1
Day 2
Day 8
Day 15

Study population

Inclusion criteria:

- Informed consent / pediatric assent, if applicable
- Male or female 10-18 years of age, inclusive
- Genetically confirmed diagnosis of PWS
- PWS nutritional phase 3 (based on Miller, 2011)
- HPWSQ-R score greater than 13 at screening
- Negative urine drug screen
- Negative pregnancy test for females of child bearing potential
- Ability and willingness to comply with protocol and trial requirements

Exclusion criteria:

- Known genetic, hormonal, or chromosomal cause of cognitive impairment other than PWS
- Presence of currently active psychotic symptoms
- Presence of cardiovascular disorders, epilepsy, frequent migraines or severe asthma
- Previous diagnosis of autism spectrum disorder by a qualified healthcare provider
- Prior or concomitant use of SSRI or SNRI, antipsychotic medication, wakefulness-promoting drug, or thyroid hormone unless the prescribed dose of medication has been stable for >= 6 months at time of screening
- Use of any of the prohibited medications or therapies listed below:
 - Intranasal therapy, including nasal saline
 - Any product containing prostaglandins
- Major surgery within 6 months of screening
- Nasal or sinus surgery within 1 year of screening
- Chronic sinusitis - more than 3 episodes per year
- Other nasal diseases that may affect deposition of intranasal medication
- Serum sodium < 135 mmol/L at screening
- Known hypersensitivity to any component of carbetocin or saline solution
- History of long term daily use of any intranasal medication
- Cancer within the last 5 years except for adequately managed basal cell carcinoma or squamous cell carcinoma of the skin
- History within the last 2 years or current abuse of drugs or alcohol
- Intake of an investigational medicinal product in the 12 weeks preceding screening or longer, if judged by the investigator to possibly influence the outcome of the trial
- Mental incapacity or language barrier of the primary parent/caregiver precluding adequate understanding or cooperation
- Considered by the investigator to be unsuitable for the trial for any other reason

Study treatment

Subjects received intranasal carbetocin or placebo 3 times per day before meals.

Study endpoints

Primary endpoint (change from baseline to Day 15):

Hyperphagia in PWS Questionnaire-Responsiveness (HPWSQ-R) score completed by subject's parent/caregiver:

The Hyperphagia in Prader-Willi Syndrome Questionnaire (HPWSQ) is an informant-based measure originally developed by Elisabeth Dykens as a comprehensive tool to examine the psychological, developmental and neurobiological correlates of hyperphagia in PWS (Dykens, 2007). HPWSQ-R included 11 questions, structured to include a 1-week recall period. The HPWSQ-R was designed to be used for repeated measures to evaluate the change in hyperphagia severity after intervention.

HPWSQ-R was a precursor to the Hyperphagia Questionnaire for Clinical Trials (HQ-CT). This study was the first clinical trial to use HPWSQ-R to evaluate a treatment intervention. HQ-CT is now more commonly used in clinical trials in subjects with PWS.

Other endpoints (change from baseline to Day 15):

Children's Yale-Brown Obsessive-Compulsive Scale (CY-BOCS):

The CY-BOCS is a clinician rated, semi-structured inventory of specific symptoms and symptom severity in pediatric obsessive-compulsive disorder (OCD) (Scahill, 1997).

Food domain of the Reiss Profile:

The food domain of the Reiss Profile consists of 7 questions that pertain to food seeking behavior. The food domain of the Reiss Profile was evaluated by Dykens, et al. (unpublished) in patients with PWS, and results showed that persons with PWS had significantly higher scores in the Reiss Profile food domain than subjects with Williams syndrome or Down syndrome at all measurement time points.

Clinical Global Impression-Improvement Scale (CGI-I):

CGI rating scales are commonly used measures of symptom severity, treatment response and the efficacy of treatments in clinical studies of subjects with psychiatric, neurological or behavioral disorders (Guy, 1976). CGI is an overall clinician-determined summary measure that takes into account all available information including knowledge of the subject's history, psychosocial circumstances, symptoms, behavior, and the impact of the symptoms on the subject's ability to function (Busner, 2007)

Hyperphagia in PWS Questionnaire-Responsiveness (HPWSQ-R) score completed by clinician (HPWSQ-R-C):

HPWSQ-R-C recorded the clinician assessment of the questions that caregivers rated in the HPWSQ-R.

Safety was evaluated throughout the study.

Results

Baseline demographics

	Carbetocin (n=17)	Placebo (n=20)
Gender		
Male, n (%)	6 (35%)	8 (40%)
Female, n (%)	11 (65%)	12 (60%)
Age, years		
Mean (range)	13.9 (10-18)	13.6 (10-18)
Race, n (%)		
Black/African American	0 (0%)	1 (5%)
White	17 (100%)	19 (95%)
BMI (kg/m²), mean (range)	25.7 (15.8-35.3)	25.7 (15.9-40.0)
HPWSQ-R Score, mean (range)	35.6 (26-49)	39.7 (26-53)
PWS Genetic Subtype		
Paternal 15q11-q13 deletion	10	14
Uniparental Disomy	7	6

Subject disposition

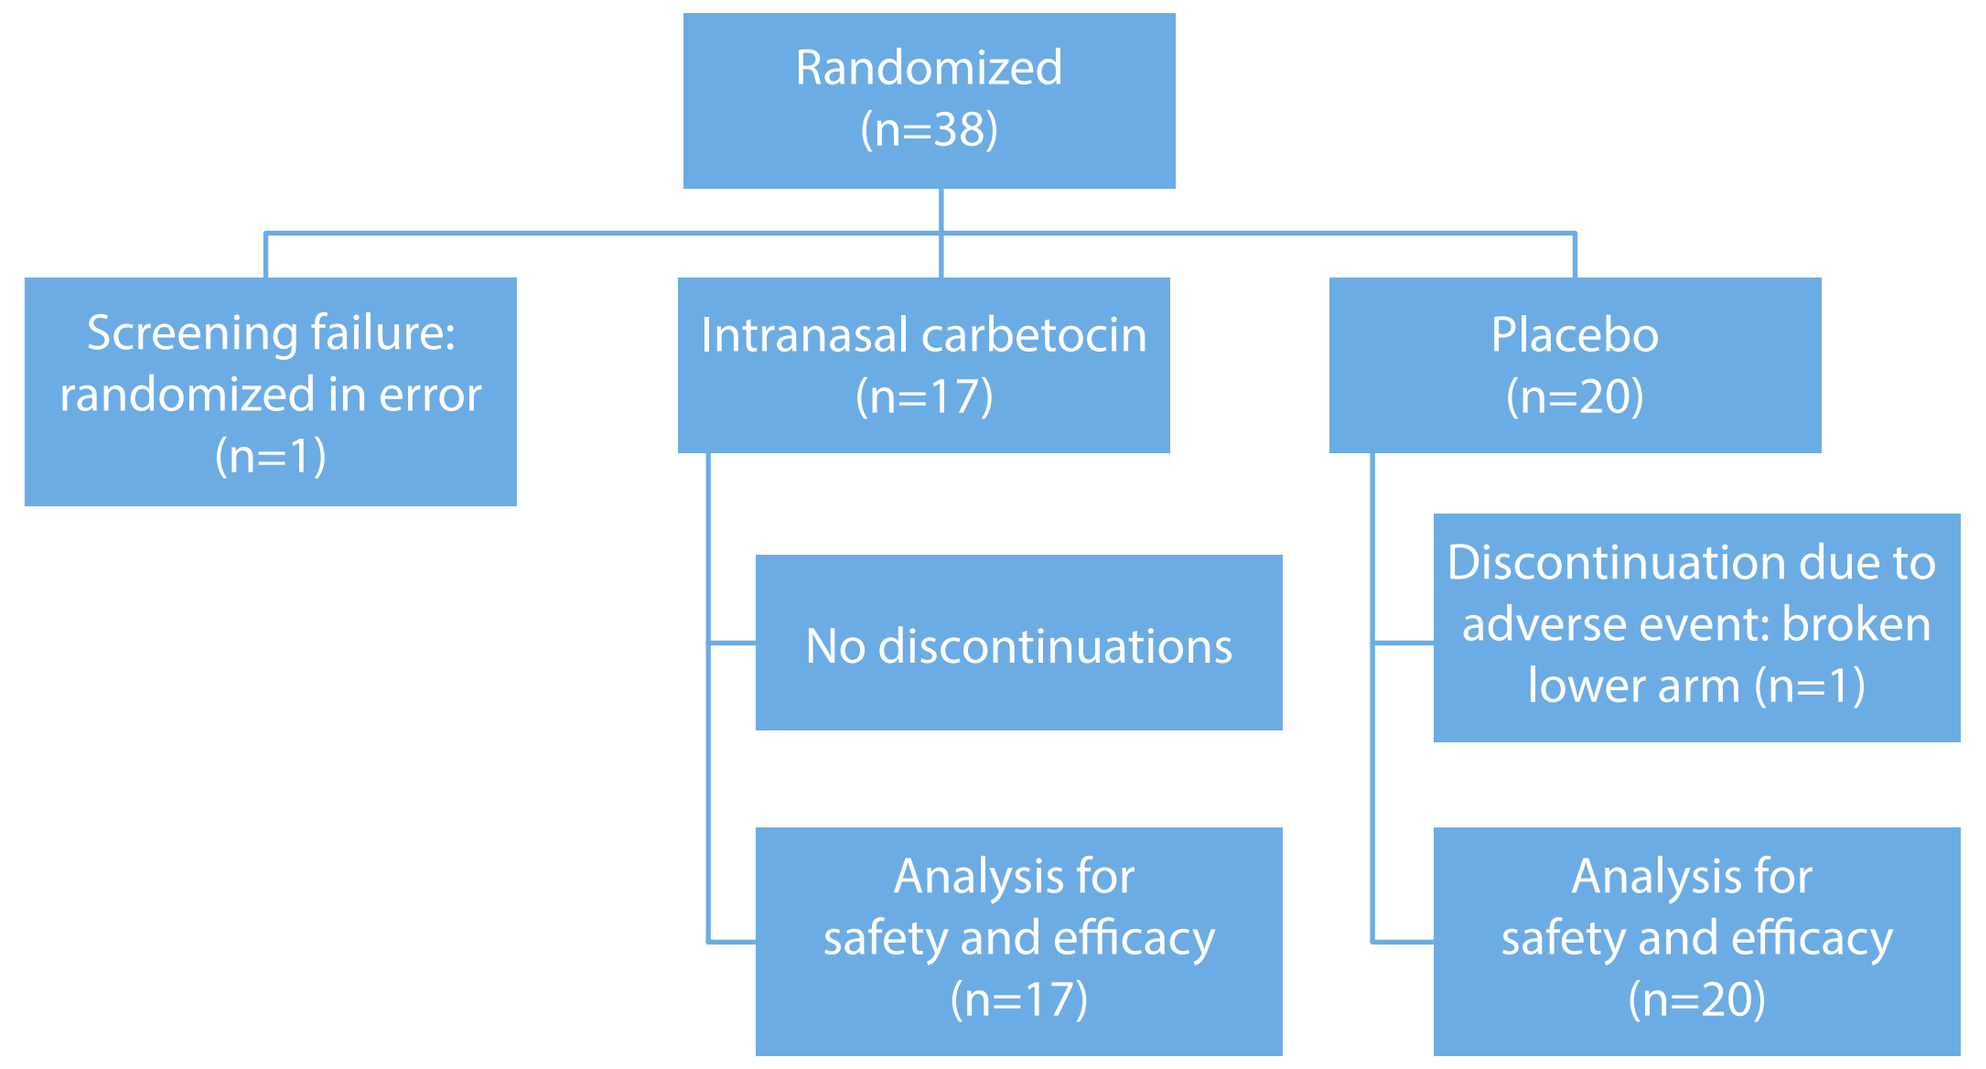
Randomized (n=38)
Screening failure: randomized in error (n=1)
Intranasal carbetocin (n=17)
Placebo (n=20)
No discontinuations
Discontinuation due to adverse event: broken lower arm (n=1)
Analysis for safety and efficacy (n=17)
Analysis for safety and efficacy (n=20)

Efficacy

Achieved pre-specified primary endpoint: HPWSQ-R at Day 15

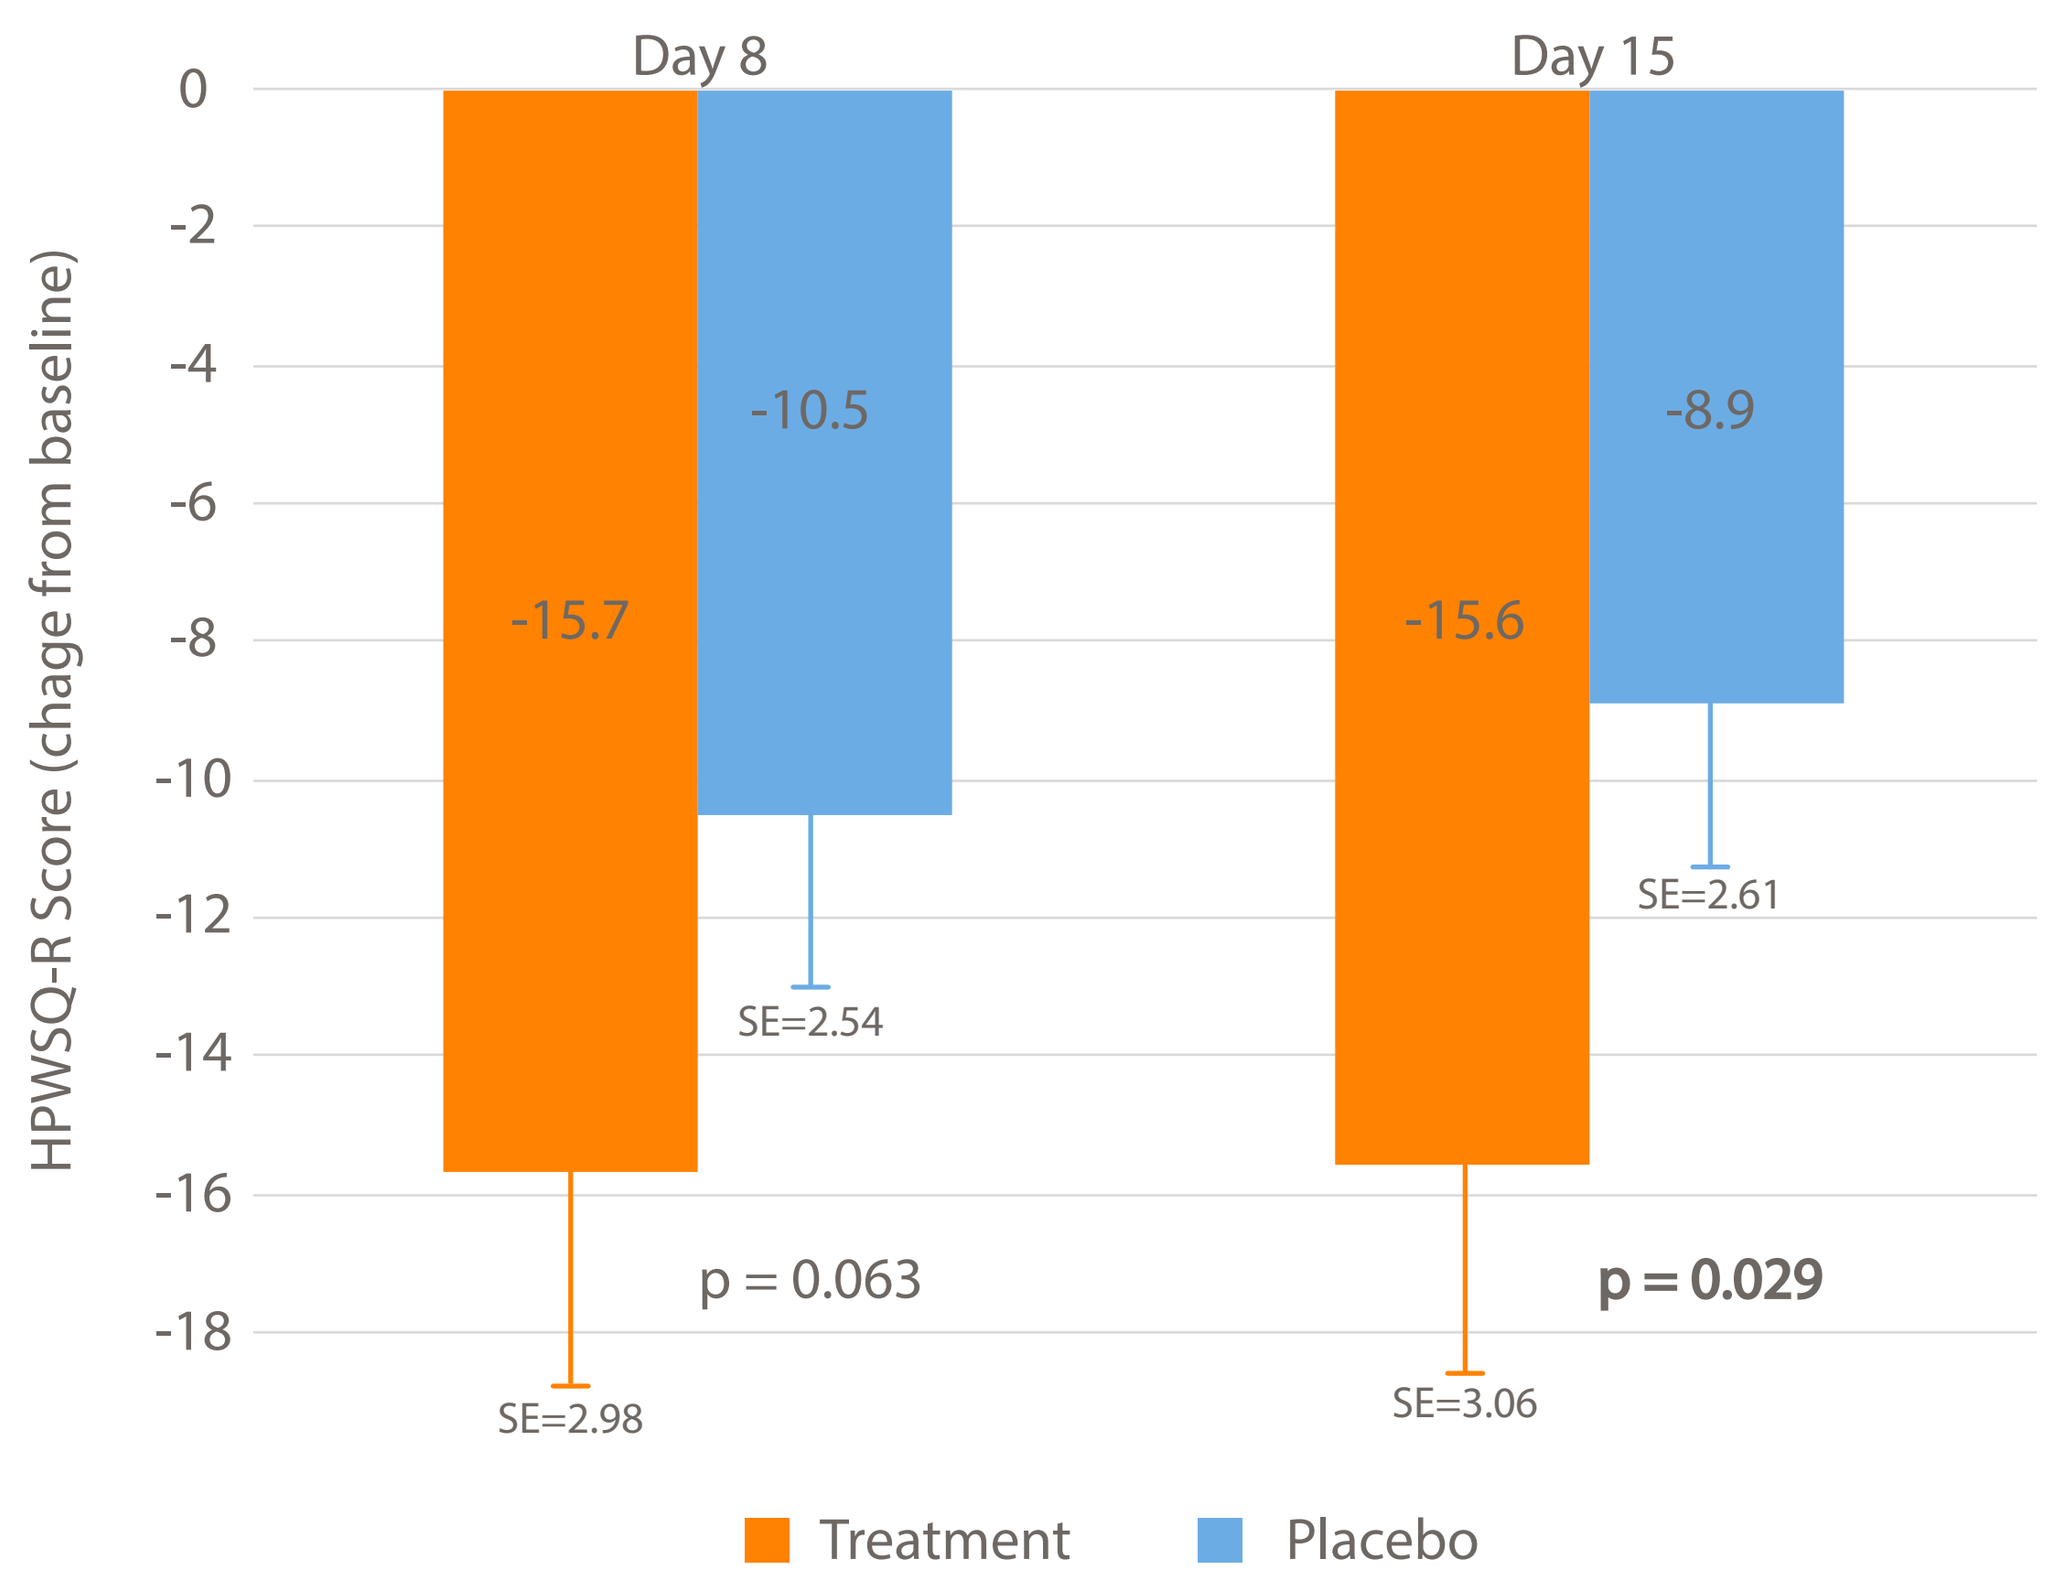
Day 8
Day 15
HPWSQ-R Score (chage from baseline)
0
-2
-4
-6
-8
-10
-12
-14
-16
-18
-15.7
-10.5
-15.6
-8.9
SE=2.98
SE=2.54
SE=3.06
SE=2.61
p = 0.063
p = 0.029
Treatment
Placebo

P-values presented are 1-sided. The 90%, 1-sided confidence interval for the -6.7 least squares mean difference excluded zero (-2.2), indicating statistical significance at the pre-specified 0.10 significance level.

Secondary endpoint efficacy results: Day 15

Efficacy Outcome	Carbetocin (n=17) LS Mean (SE) Score	Placebo (n=20) LS Mean (SE) Score	LS Mean Difference (upper limit of 90% 1-sided C.I.)	p-value (1-sided)
CY-BOCS	-8.8 (2.3)	-2.6 (1.8)	-6.2 (-3.3)	0.005
Reiss Profile Food Domain	-6.9 (1.9)	-2.5 (1.6)	-4.4 (-1.9)	0.013
CGI-I	2.7 (0.5)	3.6 (0.4)	-0.8 (-0.3)	0.023
HPWSQ-R-C Score (clinician report)	-18.9 (3.0)	-8.4 (2.5)	-10.5 (-6.3)	0.001

Safety measures

Nasal examinations: No notable trends or occurrences of nasal mucosal ulceration or perforation.
ECGs: No clinically significant abnormal ECGs.
Labs: No notable mean changes from baseline in chemistry, hematology or hemostasis values.
Vital signs: No apparent differences in vital signs values across the treatment and placebo groups.

Treatment-emergent adverse events (TEAEs)

MedDRA Preferred Term	Carbetocin (n=17)		Placebo (n=20)		Overall (n=37)	
	N (%)	E	N (%)	E	N (%)	E
Abdominal pain upper	1 (5.9)	1	1 (5.0)	1	2 (5.4)	2
Aggression	0 (0)	0	1 (5.0)	1	1 (2.7)	1
Agitation	0 (0)	0	1 (5.0)	1	1 (2.7)	1
Conjunctivitis infective	1 (5.9)	1	0 (0)	0	1 (2.7)	1
Cough	0 (0)	0	1 (5.0)	1	1 (2.7)	1
Diarrhea	1 (5.9)	1	0 (0)	0	1 (2.7)	1
Dysgeusia	1 (5.9)	9	0 (0)	0	1 (2.7)	9
Headache	5 (29.4)	8	6 (30.0)	18	11 (29.7)	26
Hyperphagia	0 (0)	0	1 (5.0)	1	1 (2.7)	1
Flushing	1 (5.9)	1	0 (0)	0	1 (2.7)	1
Medication error	1 (5.9)	1	2 (10.0)	2	3 (8.1)	3
Procedural pain	0 (0)	0	1 (5.0)	1	1 (2.7)	1
Sinusitis	0 (0)	0	1 (5.0)	1	1 (2.7)	1
Sleep disorder	1 (5.9)	1	0 (0)	0	1 (2.7)	1
Sneezing	1 (5.9)	2	0 (0)	0	1 (2.7)	2
Throat irritation	1 (5.9)	1	0 (0)	0	1 (2.7)	1
Ulna fracture	0 (0)	0	1 (5.0)	1	1 (2.7)	1
Urine analysis abnormal	0 (0)	0	1 (5.0)	1	1 (2.7)	1

N(%) = number and percentage of subjects with adverse events; E = number of adverse events reported

Discussion

Hyperphagia represents a significant unmet medical need in PWS. A reduction in hyperphagia, as measured by HPWSQ-R, was observed in patients with PWS in this Phase 2 clinical study of intranasal carbetocin. Although the HPWSQ-R results cannot be directly converted to HQ-CT, a subsequent analysis of this data with questions that correspond to HQ-CT resulted in significant reductions in scores versus placebo (LS mean difference of -5.6 from baseline to Day 15; p=0.027), demonstrating that, although the study was conducted with an older instrument, the results remain relevant using current research methods.

These results also demonstrate that hyperphagic and behavioral symptoms of PWS require broad assessment in the setting of clinical trials and clinical care. Available instruments, such as CY-BOCS and CGI, are important measures of investigational treatments, and new tools may also be warranted to assess broader treatment effects.

Conclusion

Intranasal carbetocin treatment was associated with significant reductions in HPWSQ-R total score versus placebo (LS means of -15.6 and -8.9, respectively; p=0.029), CY-BOCS (LS means of -8.8 and -2.6, respectively p=0.0047), and Reiss Profile Food domain (LS means -6.9 and -2.5, respectively; p=0.013). Intranasal carbetocin was associated with an improved clinical presentation, as measured by the CGI-I (p=0.023). These effficacy results, taken together with the safety and tolerability observed in this study, support further evaluation of intranasal carbetocin for PWS.

A Phase 3 study is planned to begin Fall 2018.

Acknowledgements

The authors acknowledge and thank the patients and families who participated in this clinical research study. The authors also thank the study team at Ferring.

References

Busner J and Targum SD. "The clinical global impressions scale: applying a research tool in clinical practice." Psychiatry (Edgmont) 2007; 4(7): 28-37
Dykens EM, et al. Assessment of Hyperphagia in Prader-Willi syndrome. Obesity. 2007;15:1816-26.
Guy W. Early Clinical Drug Evaluation (ECDEU) Assessment Manual for Psychopharmacology, Revised. National Institute of Mental Health, Rockville, MD. 1976.
Miller JL, et al. Nutritional phases in Prader-Willi syndrome. Am J Med Genet A. 2011;155:1040-9.
Scahill L, et al. Children's Yale-Brown Obsessive Compulsive Scale: Reliability and Validity. J Am Acad Child Adolesc Psychiatry. 1997;36:844–52.

Conflicts of Interest

E.M. Dykens reports grants from NIH and the Foundation for Prader-Willi Research outside of the submitted work. **J. Miller** has received research funding from Ferring, Zafgen, and Rhythm Pharmaceuticals. **M. Angulo** and **E. Roof** report that no conflicts of interest exist. **K. King** and **S. Cotter** are employees of Levo Therapeutics.